Consistency of Major Allergen Concentration and Multi-Allergen IgE-Binding Potency Across Production Lots of Protease-Rich Fungal and Insect Extracts

TJ Grier, PhD, DM LeFevre, BS, EA Duncan, BS, RS Jacobson, MS, RE Esch, PhD and TC Coyne, MD
Research & Development Laboratory Greer Laboratories, Inc. Lenoir, North Carolina USA

Abstract

Introduction
The use of non-standardized allergenic extracts is essential to the accurate diagnosis and effective treatment of allergic reactions. The quality and consistency of these products are challenged by the lack of U.S. standards of potency, genetic or climate-related variations in pollen characteristics, and cultivation differences for dust mites, fungi and insects. The high protease content associated with fungal and insect extracts poses a further risk to product reproducibility and the stability of major and minor allergens.

Methods
To examine the consistency of non-standardized allergenic products manufactured at Greer, Alternaria and German cockroach extracts were selected because they represent worst-case examples due to their endogenous proteolytic enzyme activities. Over a 2 year period, consecutive lots of final product (aqueous and glycerinated Alternaria, glycerinated German cockroach, all at 1:20 w/v) prepared with different source materials were monitored using validated quantitative immunoassays for major allergens (Alternaria Alt a 1, German cockroach Bla g 1 and Bla g 2) and total extract potencies (IgE binding to multiple allergens). SDS-PAGE provided detailed molecular profiles (fingerprints) of prominent protein constituents.

Results
A total of 9 aqueous Alternaria, 11 glycerinated Alternaria and 6 glycerinated German cockroach extract lots were included in this analysis. Alternaria product lots contained Alt a 1 allergen levels and multi-allergen IgE-binding potencies within a 2.0-fold range, and closely related to those determined day-to-day with a single extract control (15-20% CV). German cockroach product lots displayed Bla g 1 allergen, Bla g 2 allergen and multi-allergen potency levels within a narrower (1.5-fold) range. Batch-to-batch variations in source material lots had little or no impact on the major allergen content, overall IgE-binding reactivities or protein compositions of these products.

Conclusions
Alternaria and German cockroach extracts contained very consistent major allergen concentrations, IgE-binding potencies and protein profiles across multiple extractions and source material lots. Future development of characterized or standardized versions of these extracts can be facilitated using the immunochemical methods described in this study.

Materials and Methods

Allergenic extract concentrates of *Alternaria alternata* (aqueous 1:20 w/v and glycerinated 1:20 w/v) and *Blatella germanica* (German cockroach, glycerinated 1:20 w/v) manufactured at Greer over a 2 year period were obtained from released product inventories.

Most of the extracts evaluated in this study were prepared from unique combinations of source material lots. Multiple source material lots per extraction were required when production-scale manufacturing needs exceeded the yields from individual fungal or insect cultivations. All extracts except one were prepared using 2-3 source material lots.

Analytical testing was initiated in October 2010 with extracts stored for up to 14 months at 2-8°C. Subsequent testing was performed within 3 months of extract manufacturing and product lot release.

Quantitative analyses of the immunochemical compositions of these products were conducted using validated allergen-specific ELISA or multi-allergen IgE ELISA inhibition assays. Mouse monoclonal antibodies and purified references for major allergens Alternaria Alt a 1, German cockroach Bla g 1 and Bla g 2 were obtained from Indoor Biotechnologies. Stable, lyophilized reference extracts (Alternaria lot XPM1-H9, German cockroach lot XPB46-R10) were prepared at Greer. Human serum pools containing IgE specificities to a diverse group of Alternaria or cockroach allergens (lots HAM1-1 and ZE-P7, respectively) were constructed at Greer from patient sera procured from allergy clinics (HAM1-1) or plasma banks (ZE-P7).

For the ELISA procedures, microtiter plates coated with saturating levels of antibodies or allergens served to capture the target analytes, which were then detected with enzyme-conjugated antibodies and visualized using chromogenic substrates. Dose-response curves for reference and test samples met strict criteria for signal-to-background ratio, dynamic range and parallelism. Mean sample results were determined by regression for specific allergen concentrations and by parallel line bioassay for total extract potencies.

SDS-PAGE (sodium dodecyl sulfate-polyacrylamide gel electrophoresis) was performed at 0.5 µg Bradford protein loads under non-reducing conditions using 12% acrylamide gels (Bio-Rad Mini-Protean II), with protein band patterns visualized by silver staining (Invitrogen Silver Xpress).

Intra-dilutional standard deviations (SD) for each extract lot result, inter-assay SD across product lots, and coefficients of variability (% CV, = SD/Mean x 100) were also determined.

Aqueous Alternaria Extracts

The 9 aqueous Alternaria extract lots analyzed in this study were prepared from a total of 21 unique source material preparations, and exhibited Bradford protein concentrations ranging from 0.309-0.468 mg/mL (mean: 0.379; SD: 0.050; %CV: 13.2).

Alt a 1 ELISA results across these extract lots yielded consistent mean major allergen levels with intra-dilutional CVs below 15% and an inter-product lot CV of 22.6%.

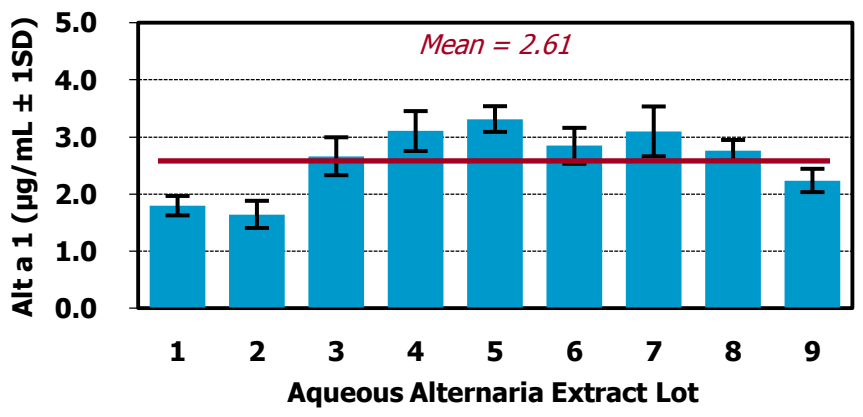
Alt a 1 (µg/mL ± 1SD)
5.0
4.0
3.0
2.0
1.0
0.0
Mean = 2.61
1 2 3 4 5 6 7 8 9
Aqueous Alternaria Extract Lot

ELISA inhibition results were also consistent across the 9 product lots, with a 22.4% inter-product lot CV, only slightly higher than the inter-assay CV of the reference lot (15.4%).

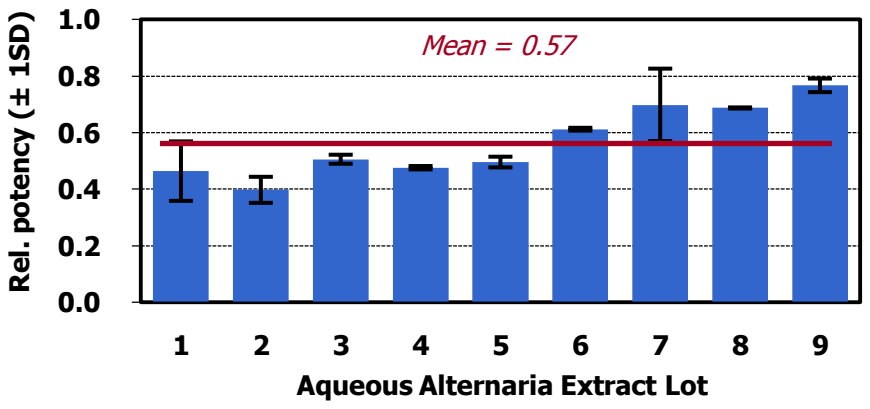
Rel. potency (± 1SD)
1.0
0.8
0.6
0.4
0.2
0.0
Mean = 0.57
1 2 3 4 5 6 7 8 9
Aqueous Alternaria Extract Lot

SDS-PAGE profiles confirmed the consistent presence of 15-20 distinct protein bands in aqueous Alternaria extracts 1-9.

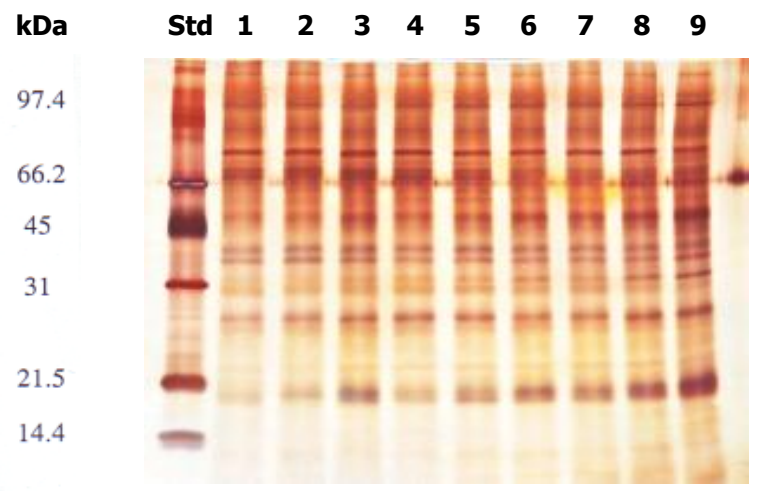
kDa
Std 1 2 3 4 5 6 7 8 9
97.4
66.2
45
31
21.5
14.4

MPN 082611H412

Consistency of Major Allergen Concentration and Multi-Allergen IgE-Binding Potency Across Production Lots of Protease-Rich Fungal and Insect Extracts

TJ Grier, PhD, DM LeFevre, BS, EA Duncan, BS, RS Jacobson, MS, RE Esch, PhD and TC Coyne, MD
Research & Development Laboratory Greer Laboratories, Inc. Lenoir, North Carolina USA

Glycerinated Alternaria Extracts

The 11 glycerinated Alternaria extract lots included in this study were prepared from 35 different cellular source material preparations, and displayed Bradford protein concentrations ranging from 0.498-0.729 mg/mL (mean: 0.615; SD: 0.080; %CV: 13.0).

Mean Alt a 1 levels in glycerinated Alternaria extracts were 15% higher than aqueous Alternaria products at the same 1:20 w/v strength, and also exhibited low intra-dilutional (< 25%) and inter-product lot (20.9%) coefficients of variability.

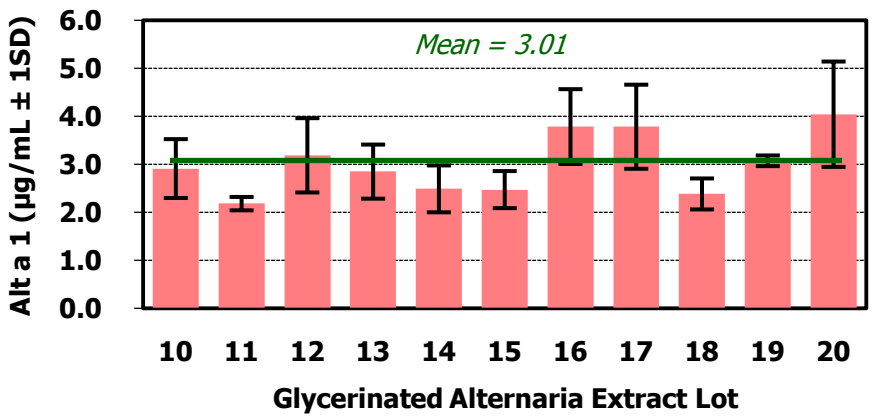
Alt a 1 (µg/mL ± 1SD)
6.0
5.0
4.0
3.0
2.0
1.0
0.0
Mean = 3.01
10 11 12 13 14 15 16 17 18 19 20
Glycerinated Alternaria Extract Lot

Mean ELISA inhibition RPs in glycerinated Alternaria products were 58% higher than in aqueous extracts, demonstrating the importance of quantifying multiple Alternaria allergens.

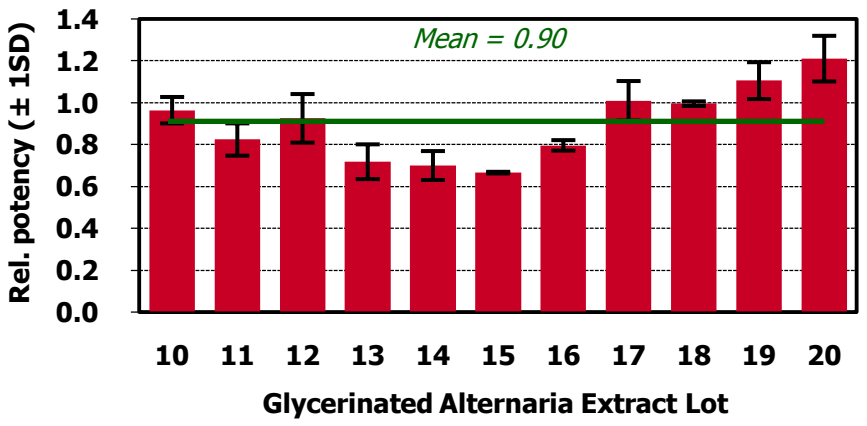
Rel. potency (± 1SD)
1.4
1.2
1.0
0.8
0.6
0.4
0.2
0.0
Mean = 0.90
10 11 12 13 14 15 16 17 18 19 20
Glycerinated Alternaria Extract Lot

SDS-PAGE band patterns were highly reproducible across glycerinated Alternaria extracts 10-20. At least 20 distinct proteins were present at similar levels in all 11 product lots.

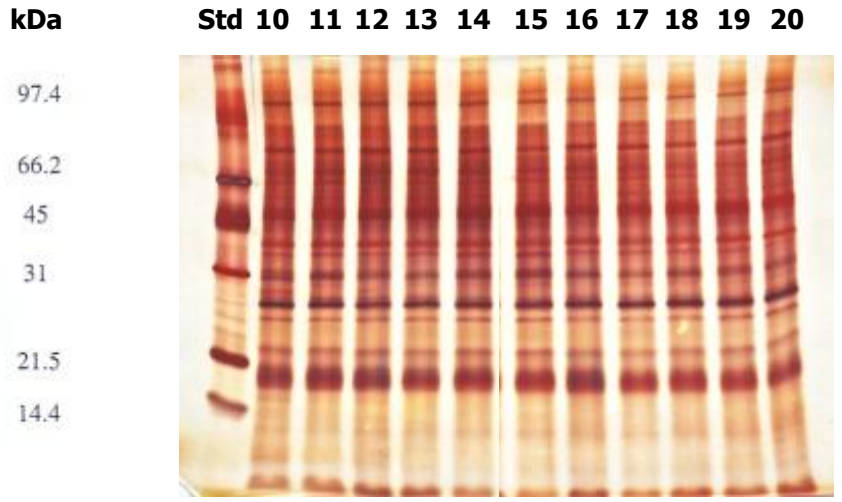
kDa
Std 10 11 12 13 14 15 16 17 18 19 20
97.4
66.2
45
31
21.5
14.4

Glyc. German Cockroach Extracts

The 6 glycerinated German cockroach extract lots included in this study were prepared from 10 different whole-insect preparations, with Bradford protein levels ranging from 0.893-1.239 mg/mL (mean: 0.997; SD: 0.141; %CV: 14.1).

Bla g 1 results displayed moderate intra-dilutional CVs (9.0-32.1%) but a lower inter-product lot CV (13.9%). Bla g 2 results were very consistent across test sample dilutions (4.6-16.2% CV) and product lots (3.5% CV).

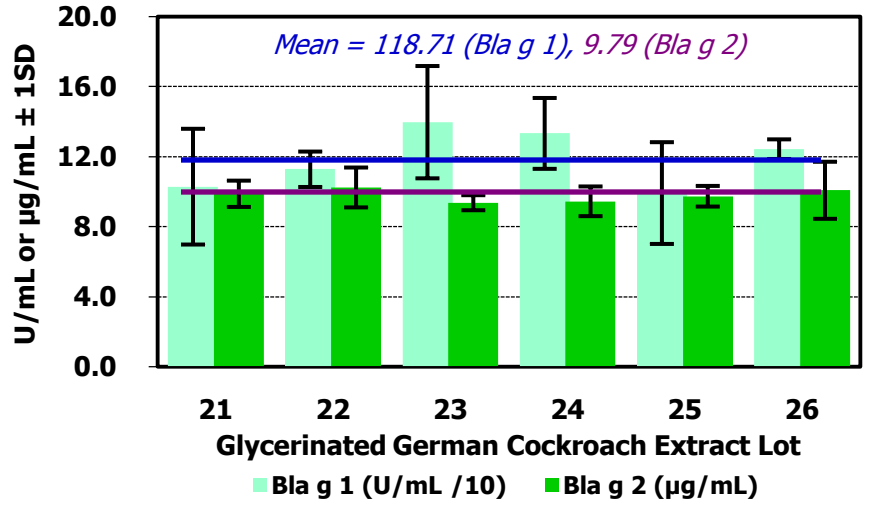
U/mL or µg/mL ± 1SD
20.0
16.0
12.0
8.0
4.0
0.0
Mean = 118.71 (Bla g 1), 9.79 (Bla g 2)
21 22 23 24 25 26
Glycerinated German Cockroach Extract Lot
Bla g 1 (U/mL /10)
Bla g 2 (µg/mL)

ELISA inhibition analysis yielded relative potency values with consistency levels (12.6% CV) comparable to replicate tests with the lyophilized German cockroach reference extract.

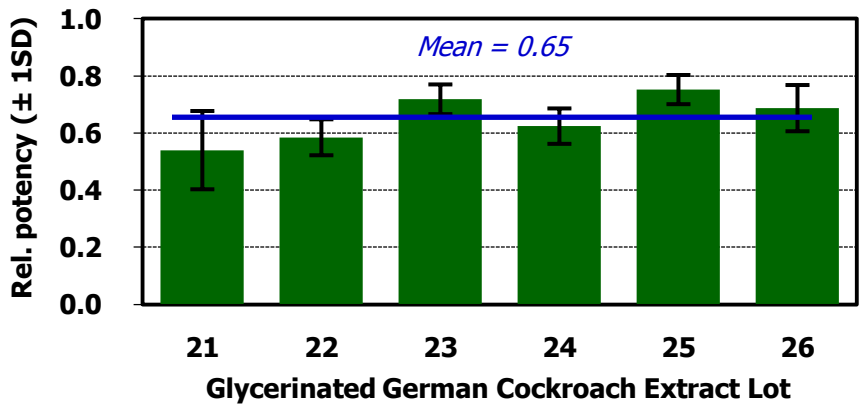
Rel. potency (± 1SD)
1.0
0.8
0.6
0.4
0.2
0.0
Mean = 0.65
21 22 23 24 25 26
Glycerinated German Cockroach Extract Lot

SDS-PAGE results confirmed the consistent presence of multiple German cockroach proteins in extract lots 21-26.

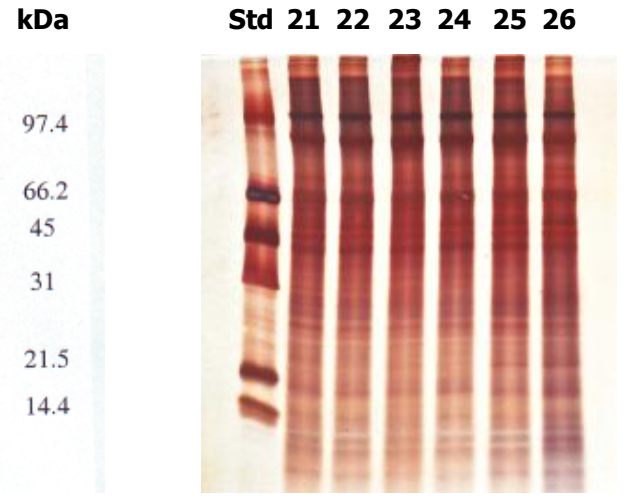
kDa
Std 21 22 23 24 25 26
97.4
66.2
45
31
21.5
14.4

Discussion

Commercial-scale production of allergenic extracts presents numerous material and process challenges to manufacturers that can directly affect the quality and consistency of product compositions, and ultimately, their clinical utility. Cultivation of fungi and insects can introduce additional sources of variability, such as differences in culture media, growth conditions, fractionation yields, and the actions of active hydrolytic enzymes during extraction and processing.

Monitoring of protease-rich fungal and insect extracts for consistency of allergen levels and IgE-binding activities across multiple product lots thus provides a meaningful worst-case study model for licensed, non-standardized extract compositions. Alternaria and German cockroach were selected for analysis based also on their overall clinical importance and prominent roles in asthma exacerbations.

The observed consistency or variability of these extracts is dependent on both the inherent reproducibility of the testing methods and actual differences in the concentrations, structures and antibody-binding activities of single (major) or multiple allergenic protein constituents.

The ELISA assays utilized in this study typically produce results within 15-20% of mean values from day-to-day and from test-to-test. The low CV values observed for both major and total allergen activities in these Alternaria and German cockroach extract lots indicate that only minor differences in allergen composition (10% or less) were detected.

Consistent representation of both major and minor allergens in these products is critical for patients presenting with varying IgE specificities to these allergens. Analysis of major allergens alone is not sufficient for patients with prominent IgE reactions to other important allergens.

Conclusions

Alternaria and German cockroach extracts prepared at Greer displayed very consistent levels of allergenic activities across multiple extraction and source material preparations.

Immunochemical monitoring of these products using sensitive, quantitative ELISA methods for both major and total allergen levels is essential to confirm the suitability of these methods as meaningful indicators of extract integrity.

MPN 082611H412